Innovations in Treatment Fidelity: Measuring Quality of Implementation for a Life Coaching and Pharmacist Counseling Intervention for Employed Diabetics

Authors
Landry Leimalani Fukunaga, MA
Denise L. Uehara, PhD

Research Objective

Treatment fidelity is a critical component in evaluating interventions to ascertain the extent that treatment staff delivered a particular intervention as intended. Aspects of treatment fidelity identified in the literature include adherence, exposure, quality of delivery, participant responsiveness and program differentiation (Dane & Schneider, 1998; Dusenbury et al., 2003). ***The objective of this study was to measure treatment fidelity within the Hawai'i Demonstration to Maintain Independence and Employment (HI-DMIE)***—a randomized controlled trial designed to investigate whether access to life coaching and pharmacist counseling could forestall or prevent the loss of employment and independence due to diabetes-related complications.

Program Components

Life Coaching

Life coaching is a collaborative process between a coach and a client with the goal of increasing motivation to maximize personal changes and professional potential (Whitmore, 2007).

Coaches assisted participants to develop a realistic and desirable action plan, provided feedback and monitored progress that helped participants stay on track. The participants took responsibility to implement the action plan (HI-DMIE, 2008).

Life Coaching included:
- Goal setting
- Online Coaching Tool
- Laptop and wireless internet

Pharmacist Counseling

Based on the Diabetes Ten City Challenge program, pharmacists provide counseling to help participants manage medications and side effects, monitor blood glucose, develop healthy diet and exercise habits, and prevent diabetes-related complications (HI-DMIE, 2009).

Pharmacists provided one-on-one consultation in a pharmacy setting to enhance understanding, increase medication adherence and encourage participants to take an active role in managing their medications (HI-DMIE, 2008).

Pharmacist Counseling included:
- Medication Therapy Management
- Motivational Interviewing

Aspects Of Treatment Fidelity Presented

Adherence	The extent to which program components, activities, and methods were delivered as intended.
Quality of Delivery	The extent to which a provider approaches a theoretical ideal in delivering program content.
Participant Responsiveness	The degree to which participants were aware of, reacted to and made recommendations about program content.

Study Design

Research Questions

1. Did life coaches and pharmacists deliver the intended program model?
2. To what extent did life coaches and pharmacists approach theoretical ideals in delivering the program?
3. How did participants perceive the delivery and effectiveness of the program?

Research Methods

Perspective Obtained	*Data Collected*	*Analysis Conducted*
• Program developers • Program staff	• Description of the program • Survey administered at the end of the intervention period	• Compared developer and staff descriptions of program • Calculated average ratings for adherence to program model
• External evaluators	• Numeric ratings of sessions that occurred throughout the intervention period*	• Calculated instrument and inter- rater reliability using intervention period data • Calculated average ratings for the individual behaviors measured, then categorized behaviors
• Treatment group participants	• Survey administered at the end of the intervention period	• Calculated average ratings for select satisfaction and perceived effectiveness measures, then categorized measures

**Life coaches and pharmacists used digital recorders to audio record meetings with treatment participants. The researchers produced numeric ratings using two Behavior Rating Scales to evaluate randomly selected recordings and their transcripts.*

Study Population

Life Coaches
Number providing service: 9

Minimum qualifications:
- Bachelor's degree in a social science

Training requirements:
- Life coach training
- Coaching by a certified executive coach
- Diabetes class and online modules
- Training for protection of human subjects
- Project data collection training

Pharmacists
Number providing service: 5

Minimum qualifications:
- Bachelor's degree in pharmacy

Training requirements:
- Certificate in Pharmaceutical Care for Patients with Diabetes
- Training in motivational interviewing
- Training on the project, research design, pharmacist roles, and data collection responsibilities

Study Limitations

- Pharmacist responses may not be representative of the sample due to low rate of survey participation
- Pharmacist counseling sessions used to assess quality of service delivery may not be representative of the entire study as sessions were only audio-recorded for part of the intervention period, due to concerns regarding how the recordings were being used

Funding Source: Centers for Medicare & Medicaid Services

Contacts: Landry Leimalani Fukunaga (lfukunag@hawaii.edu) or Denise L. Uehara (duehara@hawaii.edu)

CMS CENTERS for MEDICARE & MEDICAID SERVICES

Findings

Adherence

Background
Adherence assessed using select responses to a program staff survey
- Staff Responding to Survey
 - Life Coaches: 8 (89%)
 - Pharmacists: 2 (40%)

Summary
- When asked to define the program they delivered, the majority of staff responded with descriptions consistent with the intended model.
- When asked to rate how well they adhered to the model they just described, staff ratings were fairly high.

Consistency of Staff Definitions with Intended Model

Life Coach

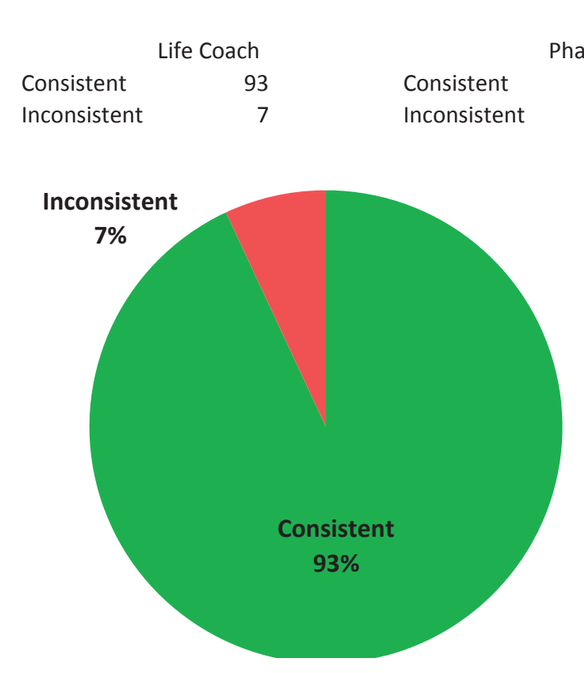

Life Coach
Consistent 93
Inconsistent 7
Inconsistent 7%
Consistent 93%

Pharmacist

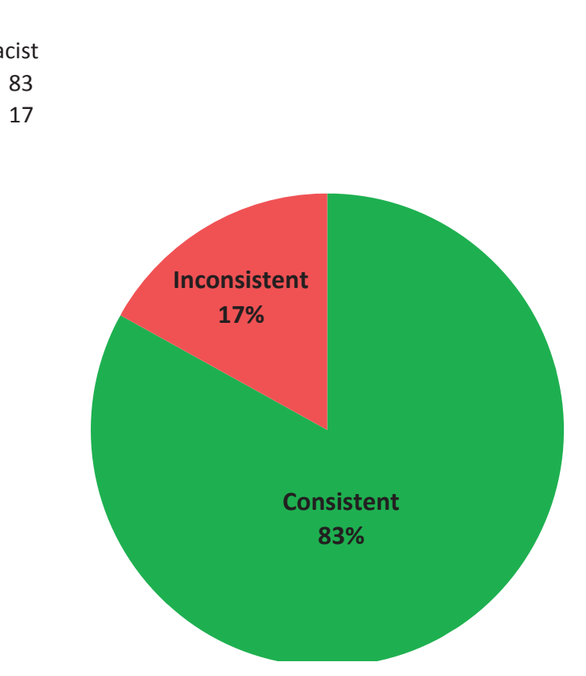

Pharmacist
Consistent 83
Inconsistent 17
Inconsistent 17%
Consistent 83%

Inconsistencies reported by staff

Life Coaches	• Ongoing modifications to the coaching model • Components of the model not used in every session • Lack of belief in a coaching model
Pharmacists	• Pharmacist required documentation not related to the HI-DMIE project

Staff Reported Model Adherence

To what degree do you feel you adhered to the model you described?	Not at all			Completely
	1	2	3	4

Pharmacists: M = 3.0, SD = 0.0
Life Coaches: M = 3.25, SD = 0.46

Quality Of Service Delivery

Background
Quality of service delivery assessed using audio recorded sessions, their transcripts, and two Behaviors Rating Scales
- Behaviors Rating Scales developed through content analysis of similar programs and collaboration with project administrators. Pilot testing took place during a feasibility study in 2007.
- 5-point rating scale (1 = needs work... 3 = basic... 5 = exemplary) across key components of each theoretical model.
- Instrument reliability
 - Life Coaching Behaviors Rating Scale: α = 0.88
 - Pharmacist Behaviors Rating Scale: α = 0.87
- All ratings conducted independently by three external evaluators
- Randomly selected audio-recorded sessions used for either:
 - Inter-rater reliability: All raters rated the same sessions
 - LC: 5 sessions rated α = 0.80
 - Ph: 3 sessions rated α = 0.79
 - Calibration throughout the rating period
 - Treatment fidelity measurement: Each session rated by one rater
 - LC: 45 sessions rated
 - Ph: 27 sessions rated

Summary
- Demonstration of key components at the basic or above basic level was considered satisfactory.
- Life coaches demonstrated all 10 key components (100%) of the life coaching model at the basic or above basic level.
- Pharmacists demonstrated 5 out of 8 key components (63%) of the pharmacist model at the basic level.

Performance by Behaviors Rating Scale Item

Life Coach (M, SD) 10 key components	***Level***	***Pharmacist (M, SD)*** 8 key components
	Exemplary (5)	
Established/Maintained Trust (4.20, 0.84) Non-judgmental (4.07, 0.94) Professionalism (3.69, 1.04) Responsive (3.56, 0.97) Coaching Relationship (3.53, 1.10)	Above Basic (4)	Maintains a Positive Environment (3.89, 1.09) Builds/Maintains Rapport (3.81, 1.14)
Asked Questions (3.36, 0.96) Active Listening (3.27, 1.07) Addressed Participant Goals (3.27, 1.19) Non Advice-giving (3.04, 1.04) Solution-Focused (2.91, 1.14)	Basic (3)	Active Listening (3.11, 0.93) Patient Centered (2.89, 0.93) Diabetes Care Monitoring (2.81, 0.92)
	Below Basic (2)	Collaborative Stance (2.44, 1.05) Goals (2.19, 1.00) Stages of Change (2.04, 1.06)
	Needs work (1)	

Performance of Theoretical Ideals

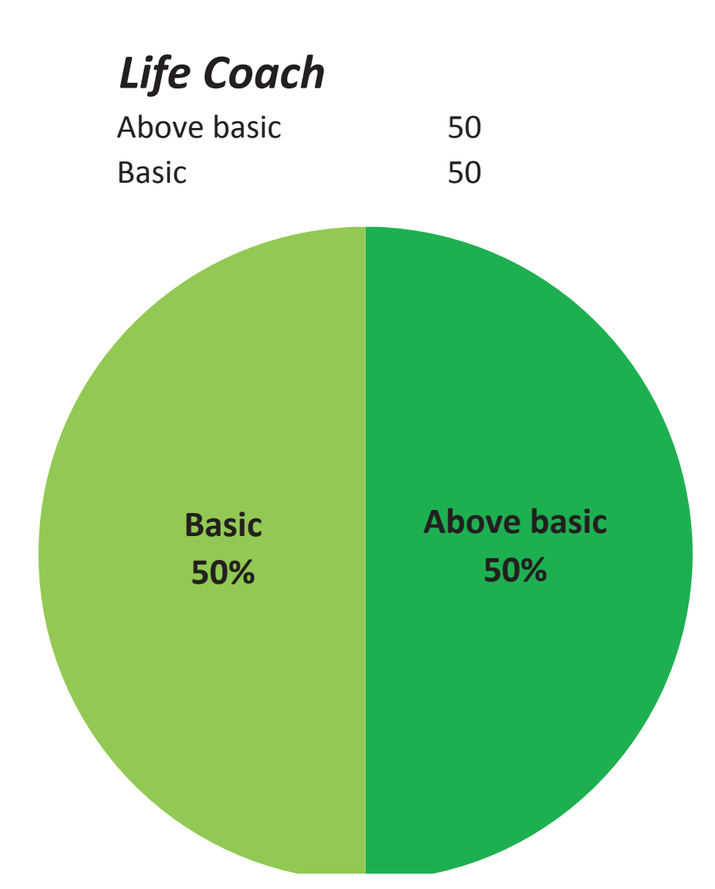

Life Coach
Above basic 50
Basic 50
Basic 50%
Above basic 50%

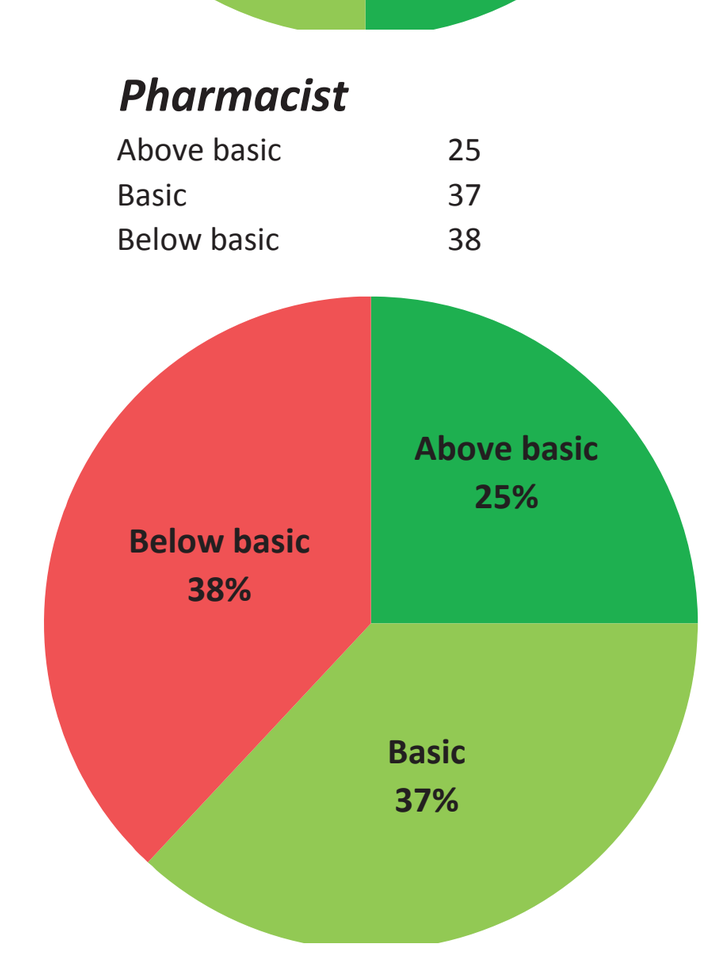

Pharmacist
Above basic 25
Basic 37
Below basic 38
Above basic 25%
Below basic 38%
Basic 37%

Participant Responsiveness

Background
Participant responsiveness assessed using Satisfaction Survey items that corresponded with Behaviors Rating Scale items
- 4-point rating scale
 - 1 = completely dissatisfied… 4 = completely satisfied
 - 1 = ineffective… 4 = very effective
- Reliability of Satisfaction Survey: α = 0.88
- Participants responding
 - Life coaching items: n = 93
 - Pharmacist counseling items: n = 86 or 85 *item dependent*

Summary
- Participants were completely satisfied with coaching behaviors and somewhat satisfied with pharmacist behaviors.
- Participants reported that coaching strategies were very effective and pharmacist strategies were somewhat effective.

Satisfaction

How satisfied are you with the following Life Coach behaviors (M, SD)	***Level***	***How satisfied are you with the following Pharmacist behaviors (M, SD)***
Professionalism (3.91, 0.38) Non-judgmental (3.90, 0.39) Coaching Relationship (3.85, 0.44) Established/Maintained Trust (3.85, 0.46) Non Advice-giving (3.83, 0.46)	Completely satisfied (4)	
	Somewhat satisfied (3)	Maintains a Positive Environment (3.17, 0.92) Builds/maintains rapport (3.16, 0.92) Diabetes Care Monitoring (3.09, 0.96) Collaborative Stance (3.08, 1.00)

Effectiveness

How effective were the following coach strategies (M, SD):	***Level***	***How effective were the following pharmacist strategies (M, SD):***
Addressed Participant Goals (3.70, 0.55) Responsive (3.68, 0.53) Solution-Focused (3.66, 0.52) Active Listening (3.60, 0.57) Asked Questions (3.58, 0.56)	Completely effective (4)	
	Somewhat effective (3)	Active Listening (2.91, 0.89) Stages of Change (2.88, 0.94) Patient Centered (2.86, 1.00) Goals (2.85, 1.01)

Conclusions

Adherence
- The majority of program staff reported adhering to the intended models.
- Inconsistent definitions of the intended models included modifications to the life coaching model after the intervention was in progress and other clinical duties required by participating pharmacies.

Quality of Delivery
- Life coaches performed all theoretical ideals at the basic or above basic level.
- Pharmacists performed a majority of the ideals at basic or above basic levels.

Participant Responsiveness
- Participants were very responsive to the life coaching component of the intervention and somewhat responsive to pharmacist counseling.

Implications for Policy, Delivery or Practice

- Measuring fidelity of a multi-component intervention was challenging but feasible.
- Defining the Life Coaching model was a complicated. Life coaching is a relatively new field with various schools of thought. Identifying specific life coach behaviors to develop the scale required a comprehensive review of the sparse literature, consulting with practicing life coaches, and working closely with project administration to determine a conceptual framework.
- Partnering with the private sector (retail pharmacies) required flexibility and patience. Pharmacists had other responsibilities and could not always provide the time to meet one on one with participants let alone effectively facilitate motivational interviewing techniques.
- Addressing concerns about confidentiality was important. Both pharmacists and life coaches were not initially comfortable with audio recording their sessions, which adversely affected data collection from pharmacists.
- Future studies should examine the extent that adherence and quality of service delivery to an intervention model affects participant satisfaction and outcomes.

References

Dane, A. V. & Schneider, B. H. (1998). Program integrity in primary and early secondary prevention: Are implementation effects out of control? Clinical Psychology Review, 18(1), 23-45.

Dusenbury, L., Brannigan, R., Falco, M., & Hansen, W. (2003). A review of research on fidelity of implementation: Implications for drug abuse prevention in school settings. Health Education Research, 18(2), 237-256.

Hawai'i Demonstration to Maintain Independence and Employment (HI-DMIE). (2008). Life Coaching Intervention Summary Review. Honolulu, HI: University of Hawaii at Manoa, Center on Disability Studies.

Hawai'i Demonstration to Maintain Independence and Employment (HI-DMIE). (2009). Live Healthy… Work Well Program Theory. Honolulu, HI: University of Hawaii at Manoa, Center on Disability Studies.

Whitmore, J. (2007). Coaching for performance: Growing people, performance and purpose (3rd ed.). Boston, MA: Nicholas Brealey Publishing.

COLLEGE OF EDUCATION UNIVERSITY OF HAWAI'I AT MĀNOA

cds CENTER ON DISABILITY STUDIES

UNIVERSITY OF HAWAI'I MĀLAMALAMA 1907 UA MAU KE EA O KA 'ĀINA I KA PONO